CUE-102 Immuno-STATs for selective targeting and expansion of WT1-specific T cells for the treatment of HLA-A02 and/or HLA-A24 cancer patients expressing WT1

Dharma Thapa, Alex Histed, Jonathan Soriano, Luke Witt , Zohra Merazga, Natasha Girgis, Miguel Moreta, Fulvio Diaz, Fan Zhao, Melissa Kemp, Paige Ruthardt, Anish Suri, Steven Quayle, John Ross and Saso Cemerski

[1]Cue Biopharma, Cambridge, Massachusetts

Background

WT1 has been ranked first amongst 75 tumor associated antigens in an effort by the National Cancer Institute to prioritize cancer antigens for therapeutic targeting. Development of novel modalities to target WT1 provide a significant opportunity to address high unmet medical need in WT1-positive malignancies, including AML, ovarian, endometrial, breast, lung, colorectal and pancreatic cancer.

We have developed novel fusion proteins, termed Immuno-STATsTM *(Selective Targeting and Alteration of T cells)*, that are comprised of HLA-A*02 or HLA-A*24 molecules presenting peptide epitopes derived from WT1. Each CUE-102 Immuno-STAT molecule also contains four copies of affinity-attenuated human interleukin-2, and an effector attenuated human immunoglobulin G Fc domain. We present here the biochemical characterization of these molecules and their bioactivity across a variety of in vitro and ex vivo studies, demonstrating the potential for CUE-102 Immuno-STATs to enhance anti-tumor immunity in patients with WT1-positive malignancies.

Cue Biopharma's Immuno-STAT Platform

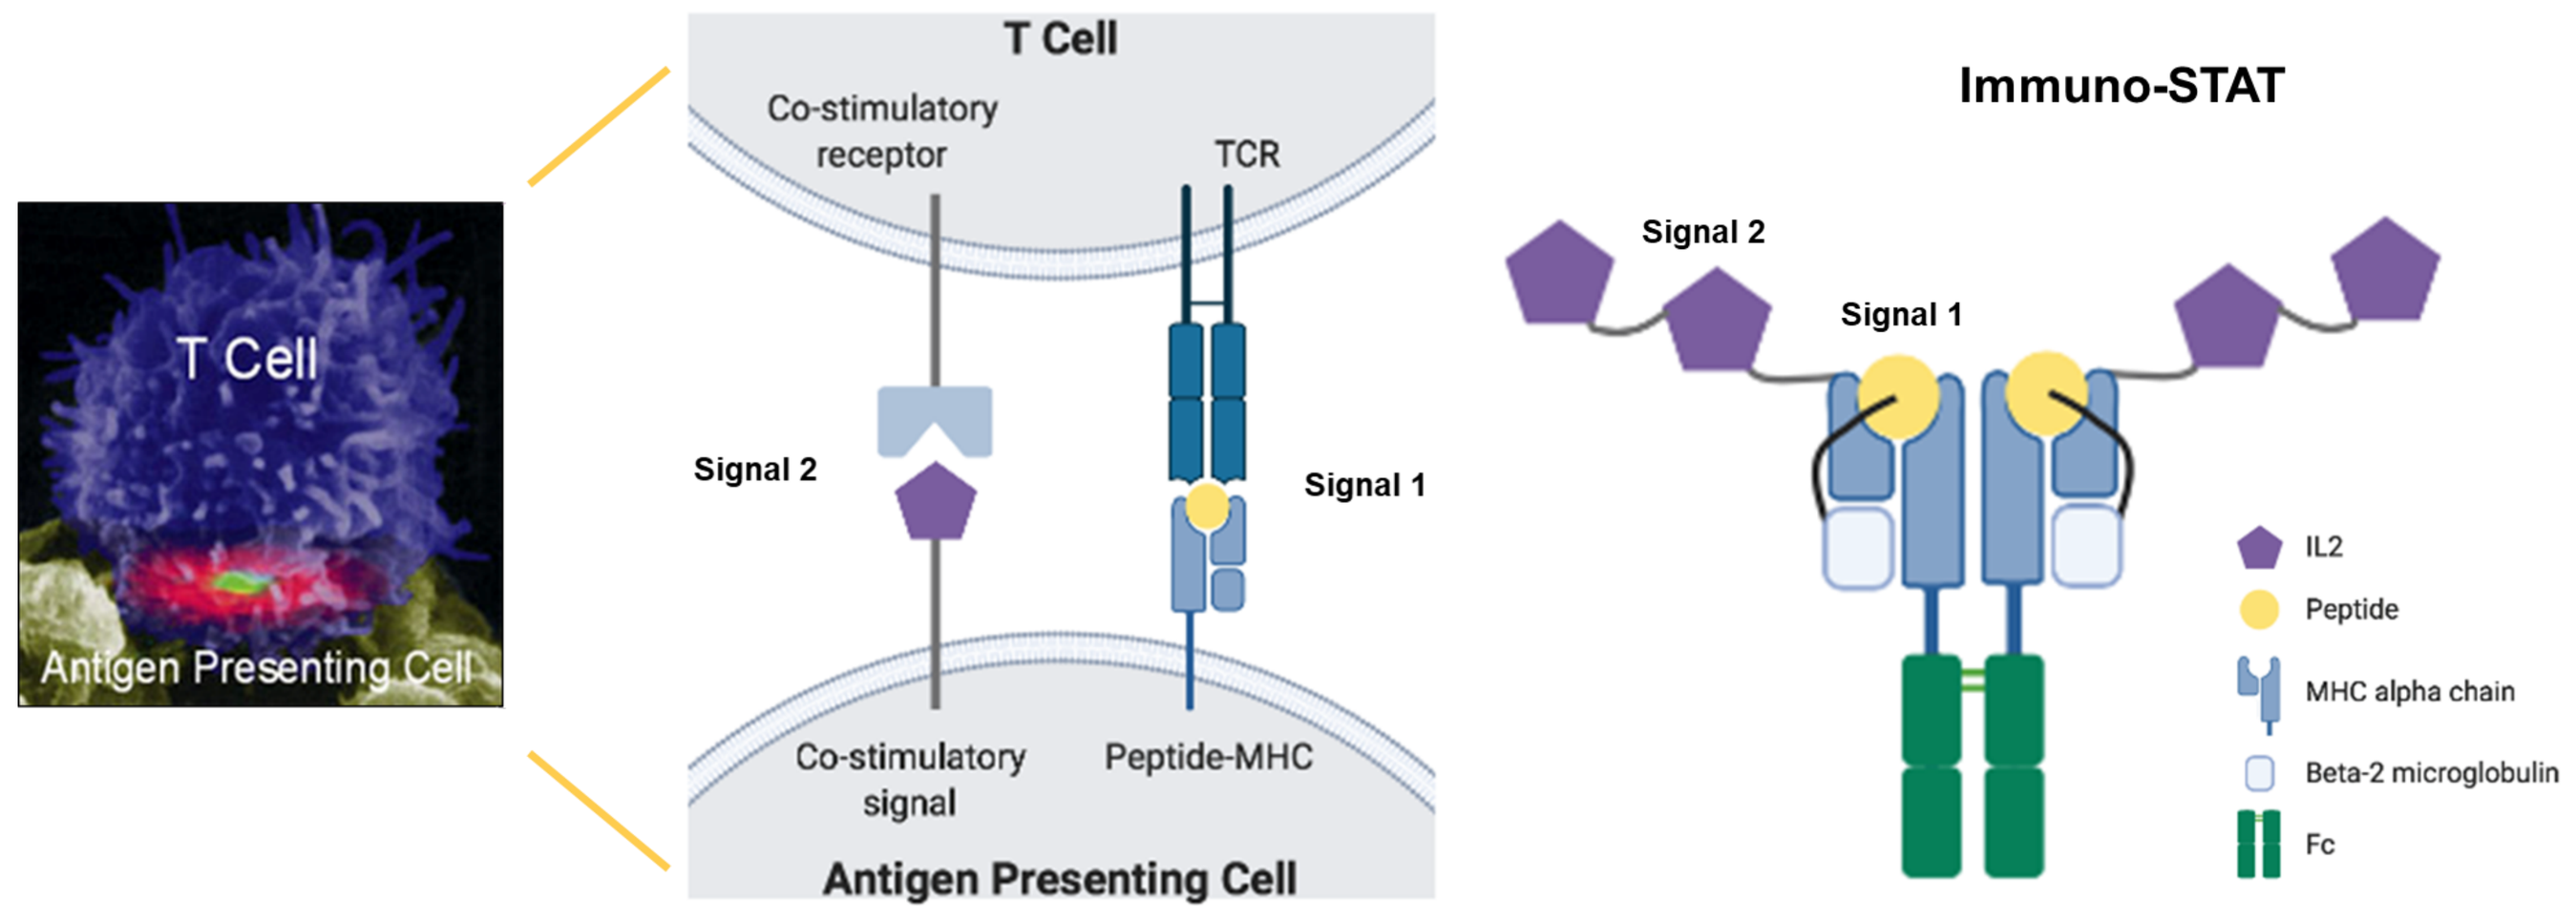

T Cell
Antigen Presenting Cell
T Cell
Co-stimulatory receptor
TCR
Signal 2
Signal 1
Co-stimulatory signal
Peptide-MHC
Antigen Presenting Cell
Immuno-STAT
Signal 2
Signal 1
IL2
Peptide
MHC alpha chain
Beta-2 microglobulin
Fc

The intimate interactions between the T cells and APCs occur within a molecular interface known as the immunological or immune synapse. The immune synapse allows controlled engagement and selective activation of T cells through the presentation of two key distinct signals: Signal 1, TCR engagement by the pMHC; and Signal 2, co-stimulatory or co-inhibitory signals. Through rational protein engineering, we have developed a proprietary class of biologics termed **Immuno-STATs** that induce and modulate T cell activity via delivery of the distinct signals provided naturally to T cells within the immune synapse. We accomplish this by the co-engineering of a TCR targeting pMHC with co-stimulatory signaling molecules in a singular biologic on an Fc framework.

Modularity of the Immuno-STAT Platform

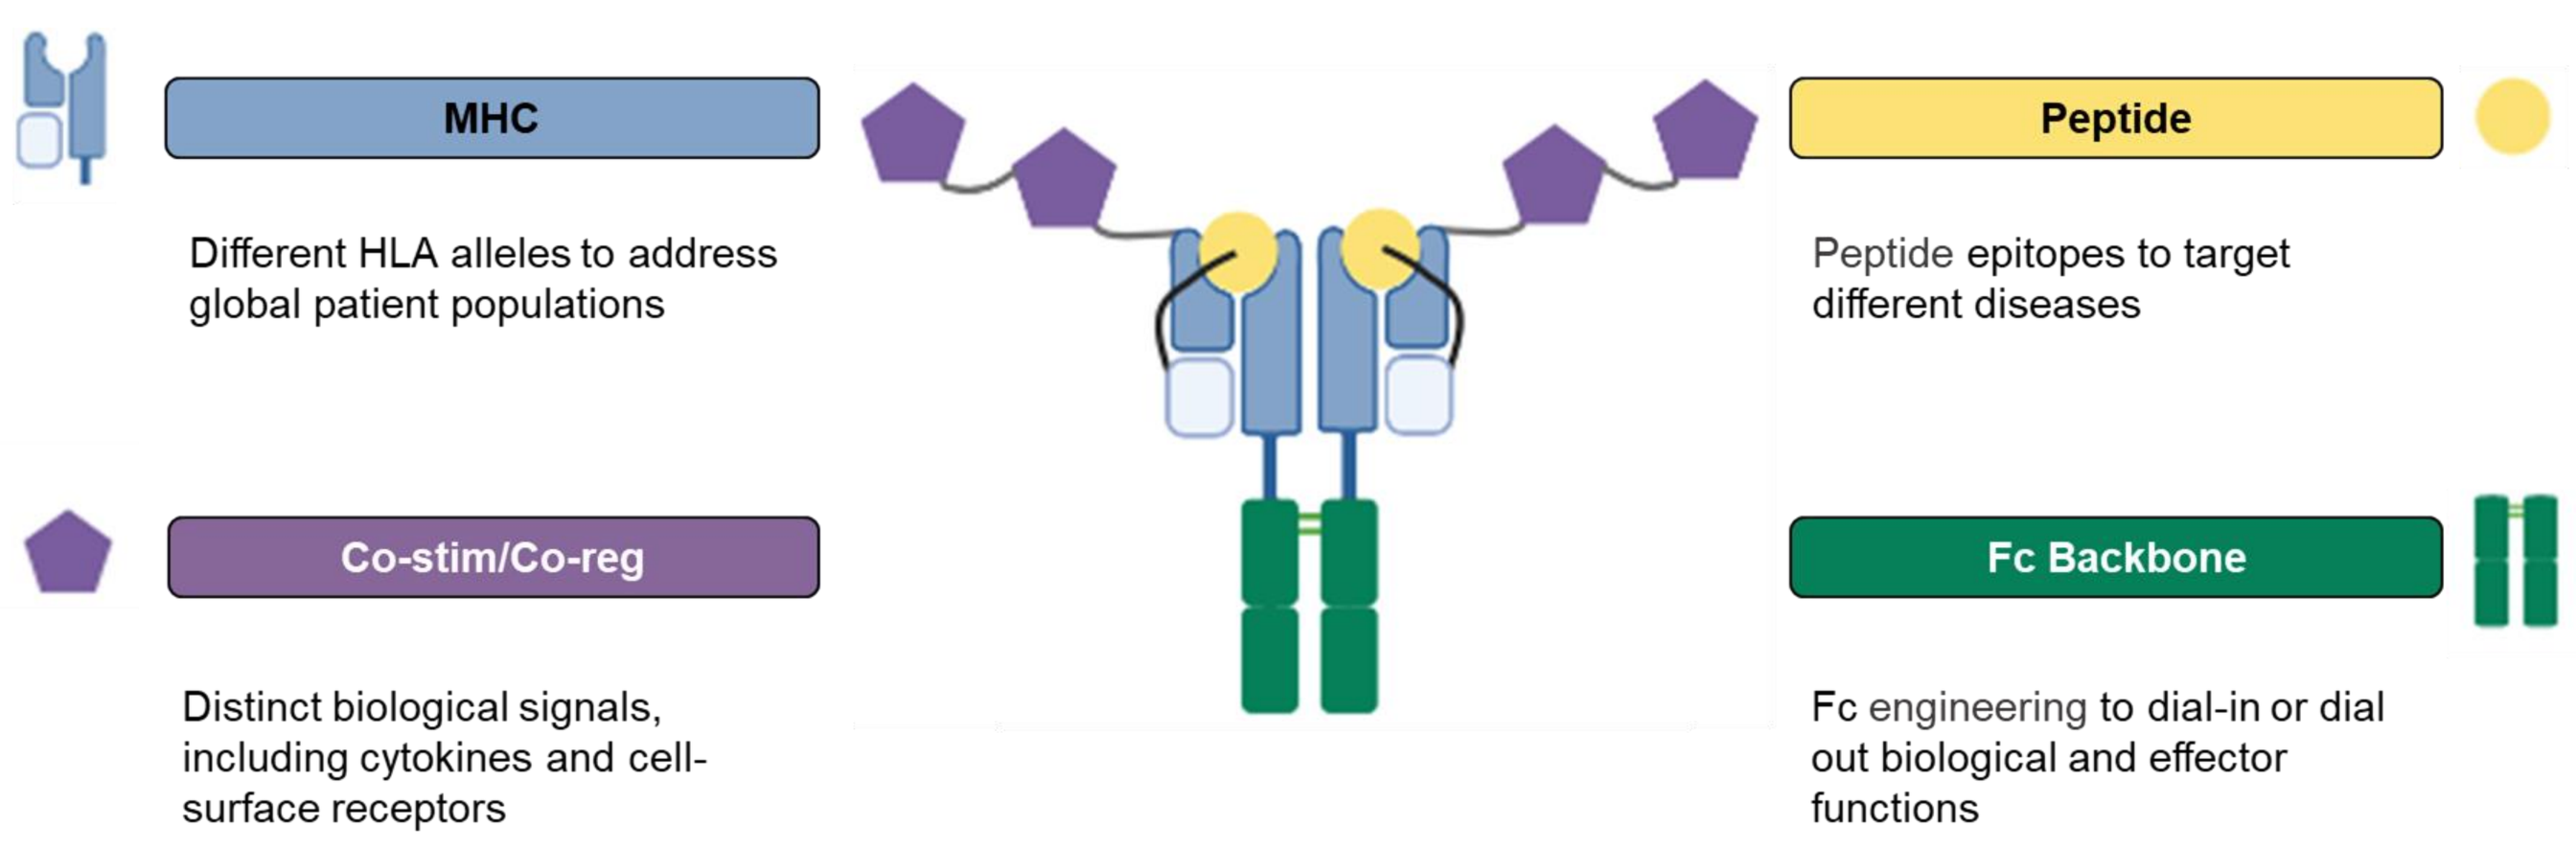

MHC
Different HLA alleles to address global patient populations
Co-stim/Co-reg
Distinct biological signals, including cytokines and cell-surface receptors
Peptide
Peptide epitopes to target different diseases
Fc Backbone
Fc engineering to dial-in or dial out biological and effector functions

All Immuno-STAT components can be readily exchanged and tailored to appropriately target and regulate the activity of disease relevant T cells in the right patient population in the right disease indication. This modularity and versatility of the platform allows us to design Immuno-STATs that address particular T cell modulation objectives/criteria, including different co-stimulatory/co-inhibitory signals or different targeting peptides and HLA alleles.

- In the context of cancer and chronic infectious disease, Immuno-STATs are designed to selectively activate disease-specific T cells
- For the treatment of autoimmune disease, Immuno-STATs are designed to selectively inhibit pathogenic autoreactive T cells

CUE-100 Series Immuno-STATs Deliver Affinity Attenuated IL-2 to Targeted Antigen-Specific T Cells

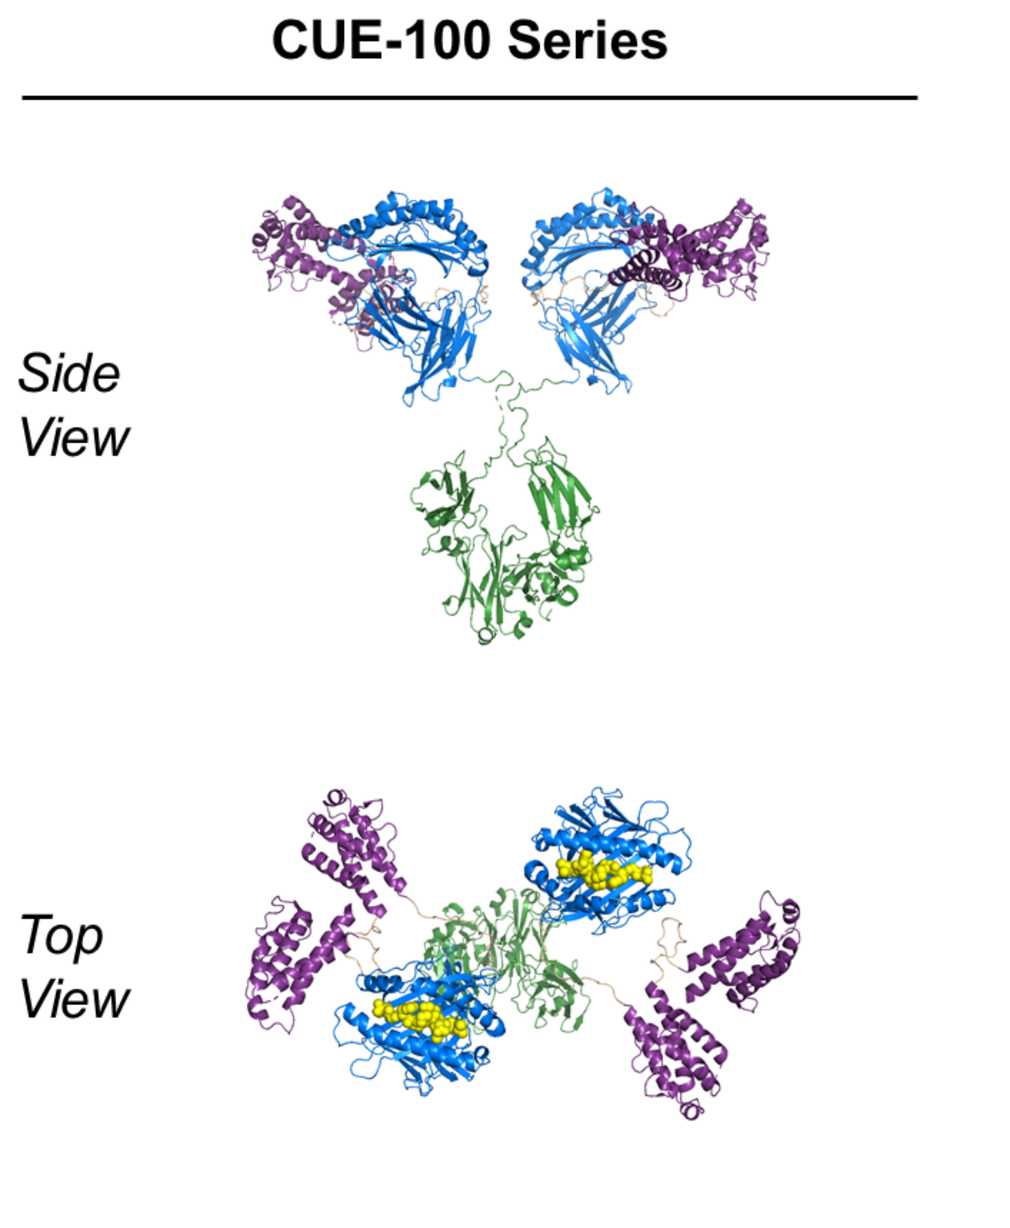

CUE-100 Series
Side View
Top View

Immune Signaling Components

Attenuated IL-2

Optimized for desired biological activity through two single amino acid changes
- Abrogates binding to IL-2R alpha subunit
- Reduces binding affinity to IL-2R beta

Maintains IL-2 ability to stimulate antigen-specific CD8+ T cells while reducing Treg expansion

Peptide Loaded HLA

Stabilized peptide HLA complex to present disease-relevant epitope to T cell receptor
- Framework allows for incorporation of an array of HLA Class I alleles (i.e., A02, A11, A24)

Provides "Signal 1" to the targeted antigen-specific CD8+ T cells, thereby enhancing the activity of the attenuated IL-2

Therapeutic framework is not dependent on barriers of antigen processing & presentation, and is designed to avoid systemic immune activation

CUE-102/A02: Design, Manufacturability Assessment and Biophysical Characterization

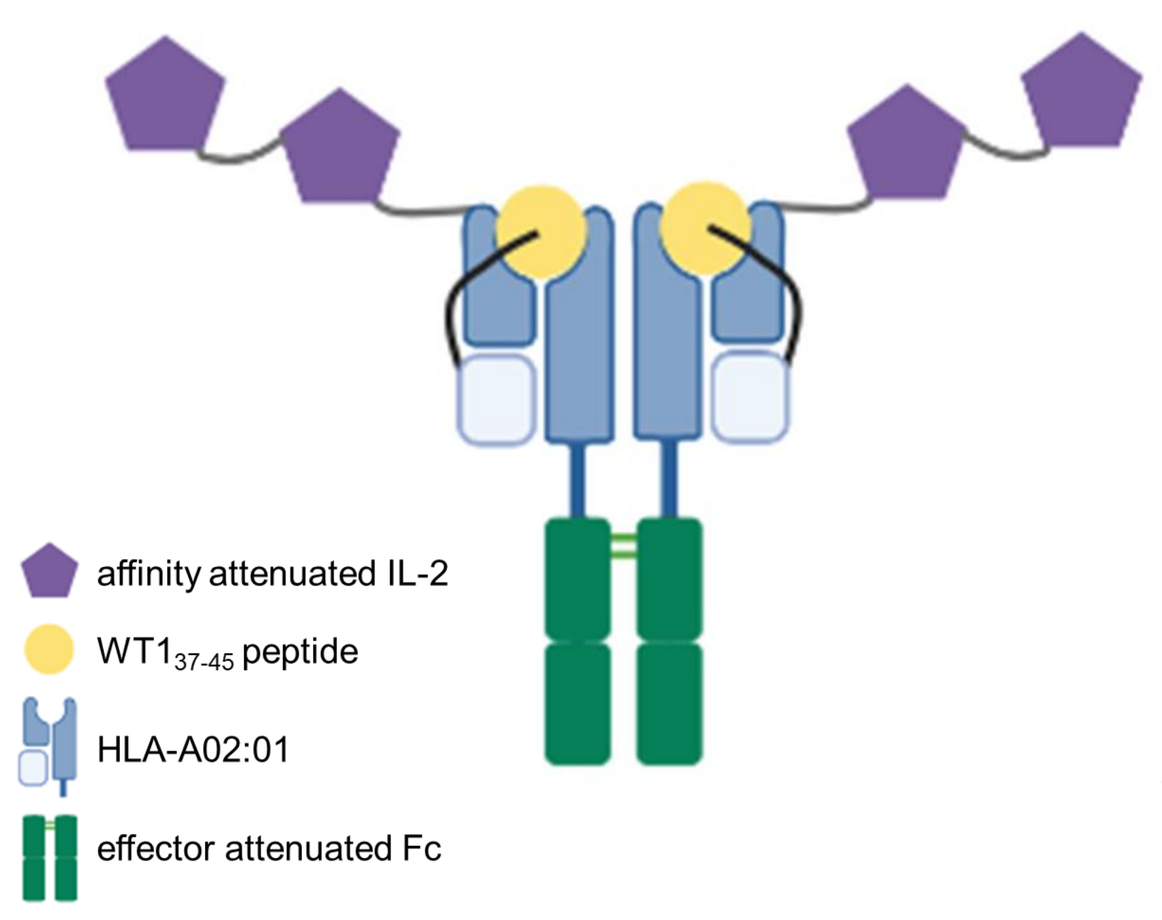

affinity attenuated IL-2
WT1 37-45 peptide
HLA-A02:01
effector attenuated Fc

Peptide	37-45
Peptide Sequence	VLDFAPPGA
Titer (mg/L)	281
% Monomer (post Protein A)	61
% Monomer (post SEC)	98
Stability (10d @ 4°C / 37°C / 42°C) Monomer Recovery %	100 / 88 / 24
Freeze-thaw (0x / 1x / 3x) % monomer by SEC	98 / 98 / 98
T_m (°C)	51
	66
	81
Intact mass (LC-MS)	Confirmed

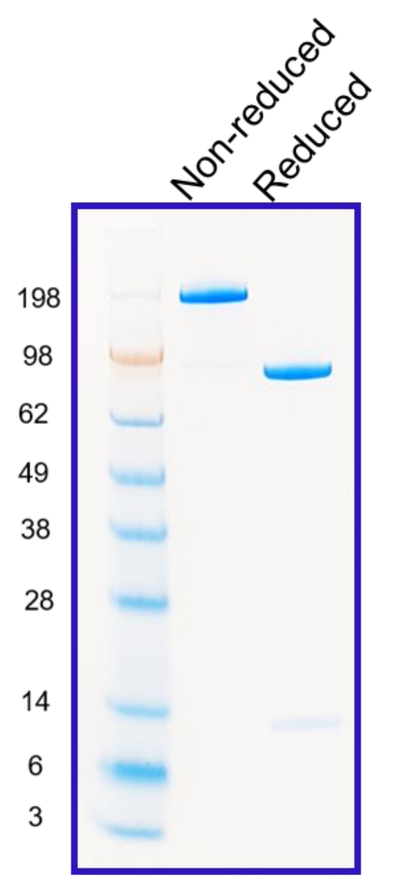

Non-reduced
Reduced
198
98
62
49
38
28
14
6
3

Diagram of CUE-102/A02. The chains are assembled (N-term to C-term) in the following order: Chain A, IL2–HLA alpha chain–Fc; Chain B, peptide–beta-2-microglobulin. Table: Manufacturability assessment and biophysical characterization of CUE-102/A02. Above: Reduced and non-reduced Coomassie-stained SDS-PAGE of the purified CUE-102/A02.

CUE-102/A02 $WT1_{37-45}$ IST Induces Expansion of $WT1_{37-45}$-Specific $CD8^+$ T Cells from Unprimed PBMCs

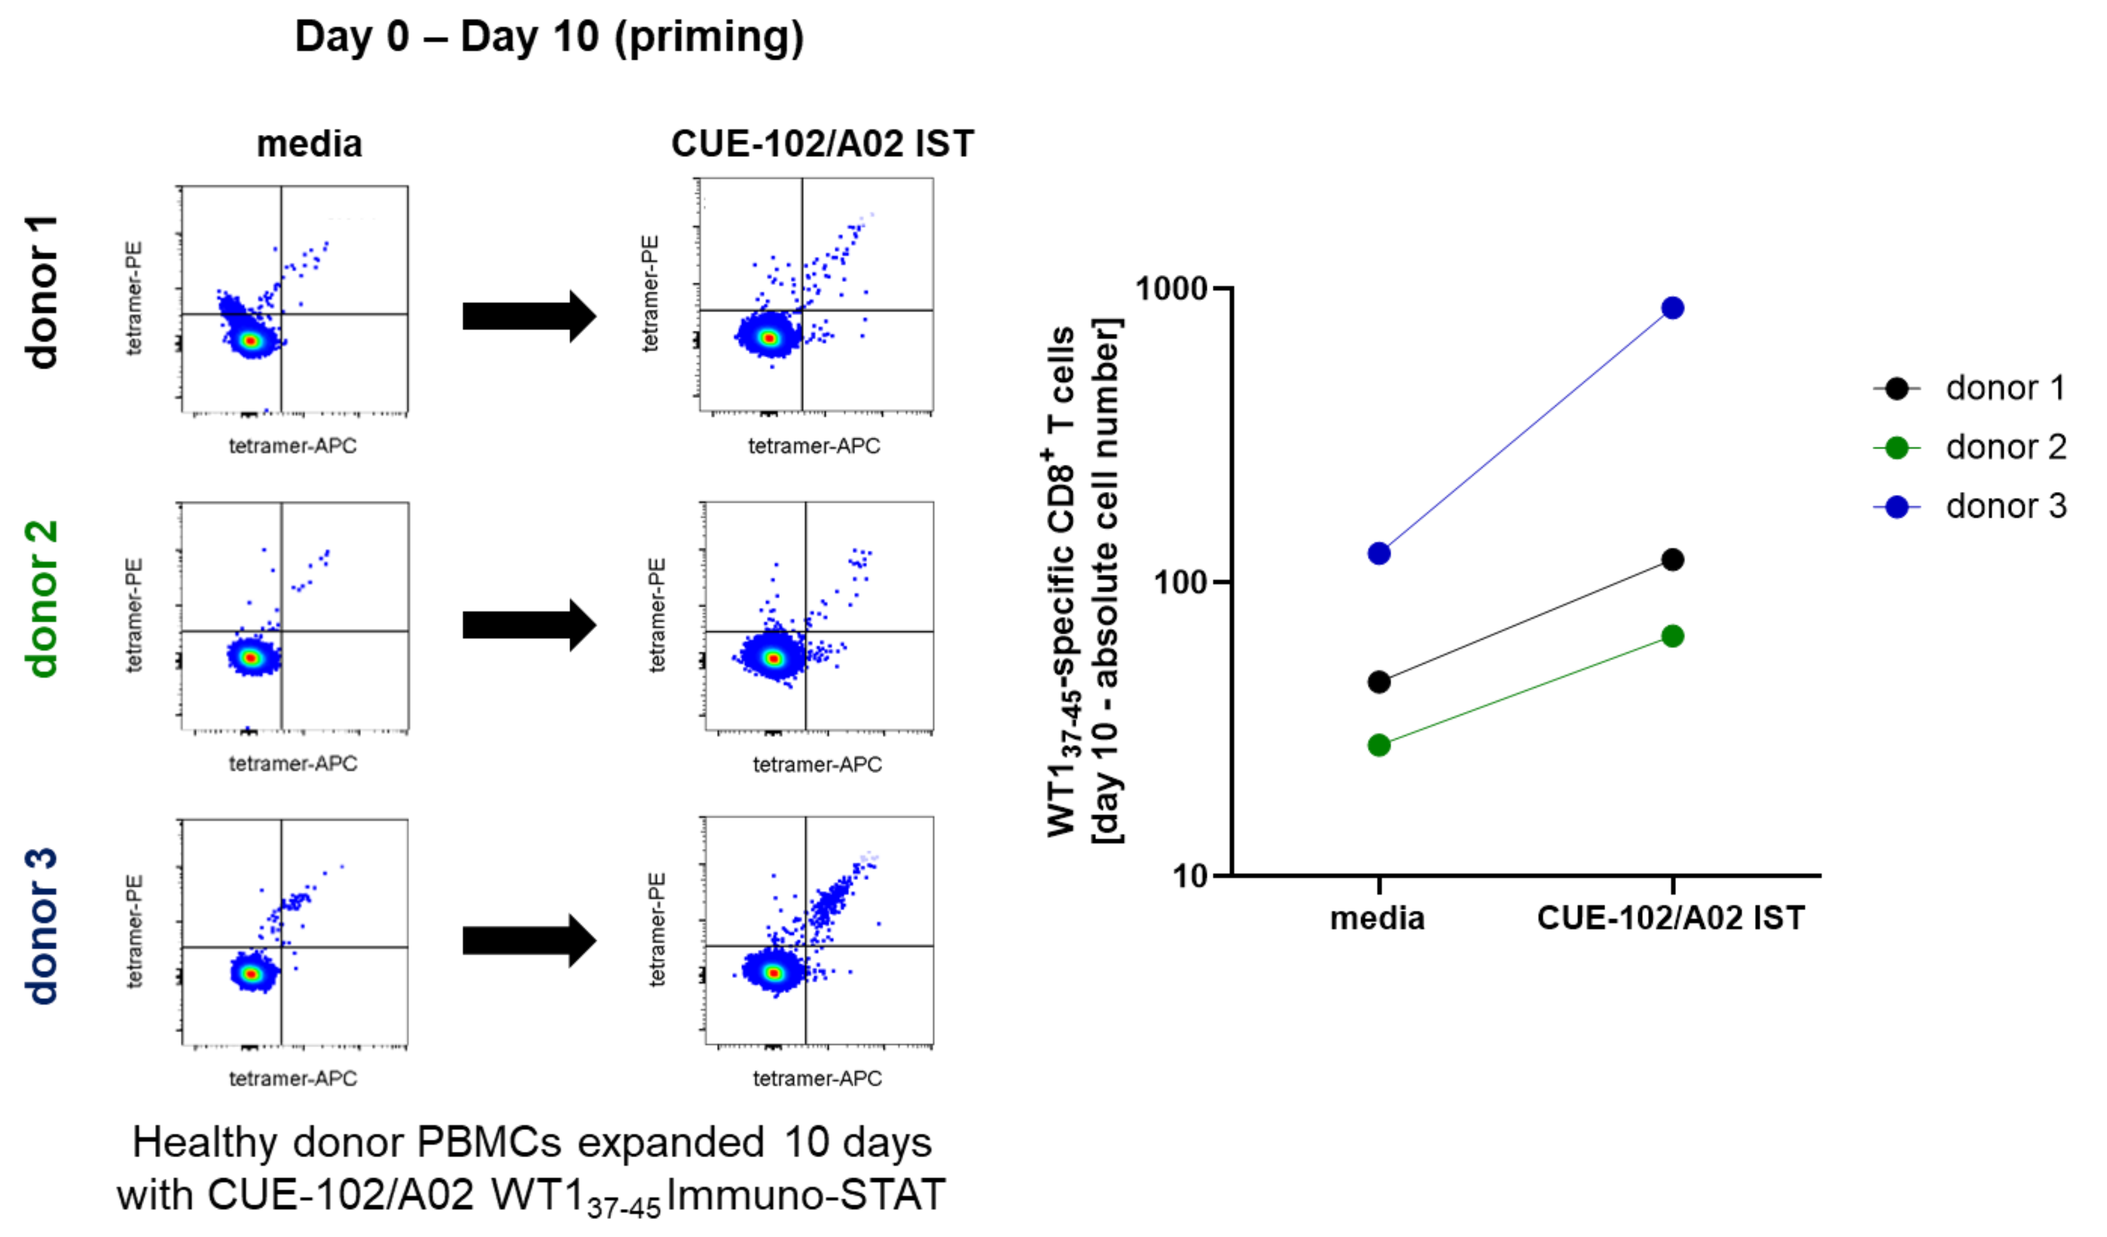

Day 0 – Day 10 (priming)
media
CUE-102/A02 IST
donor 1
donor 2
donor 3
tetramer-PE
tetramer-APC
WT1 37-45-specific CD8+ T cells [day 10 - absolute cell number]
1000
100
10
media
CUE-102/A02 IST
donor 1
donor 2
donor 3

Healthy donor PBMCs expanded 10 days with CUE-102/A02 $WT1_{37-45}$ Immuno-STAT

Healthy donor PBMCs were stimulated for 10 days with the CUE-102/A02 $WT1_{37-45}$ Immuno-STAT in ImmunocultTM media. Unstimulated cells were used as negative control. Peptide-specific $CD8^+$ T cells were detected by flow cytometry upon staining with $WT1_{37-45}$-specific tetramers.

CUE-102/A02 Immuno-STAT Induces Expansion of $WT1_{37-45}$-Specific $CD8^+$ T Cells from Primed PBMCs

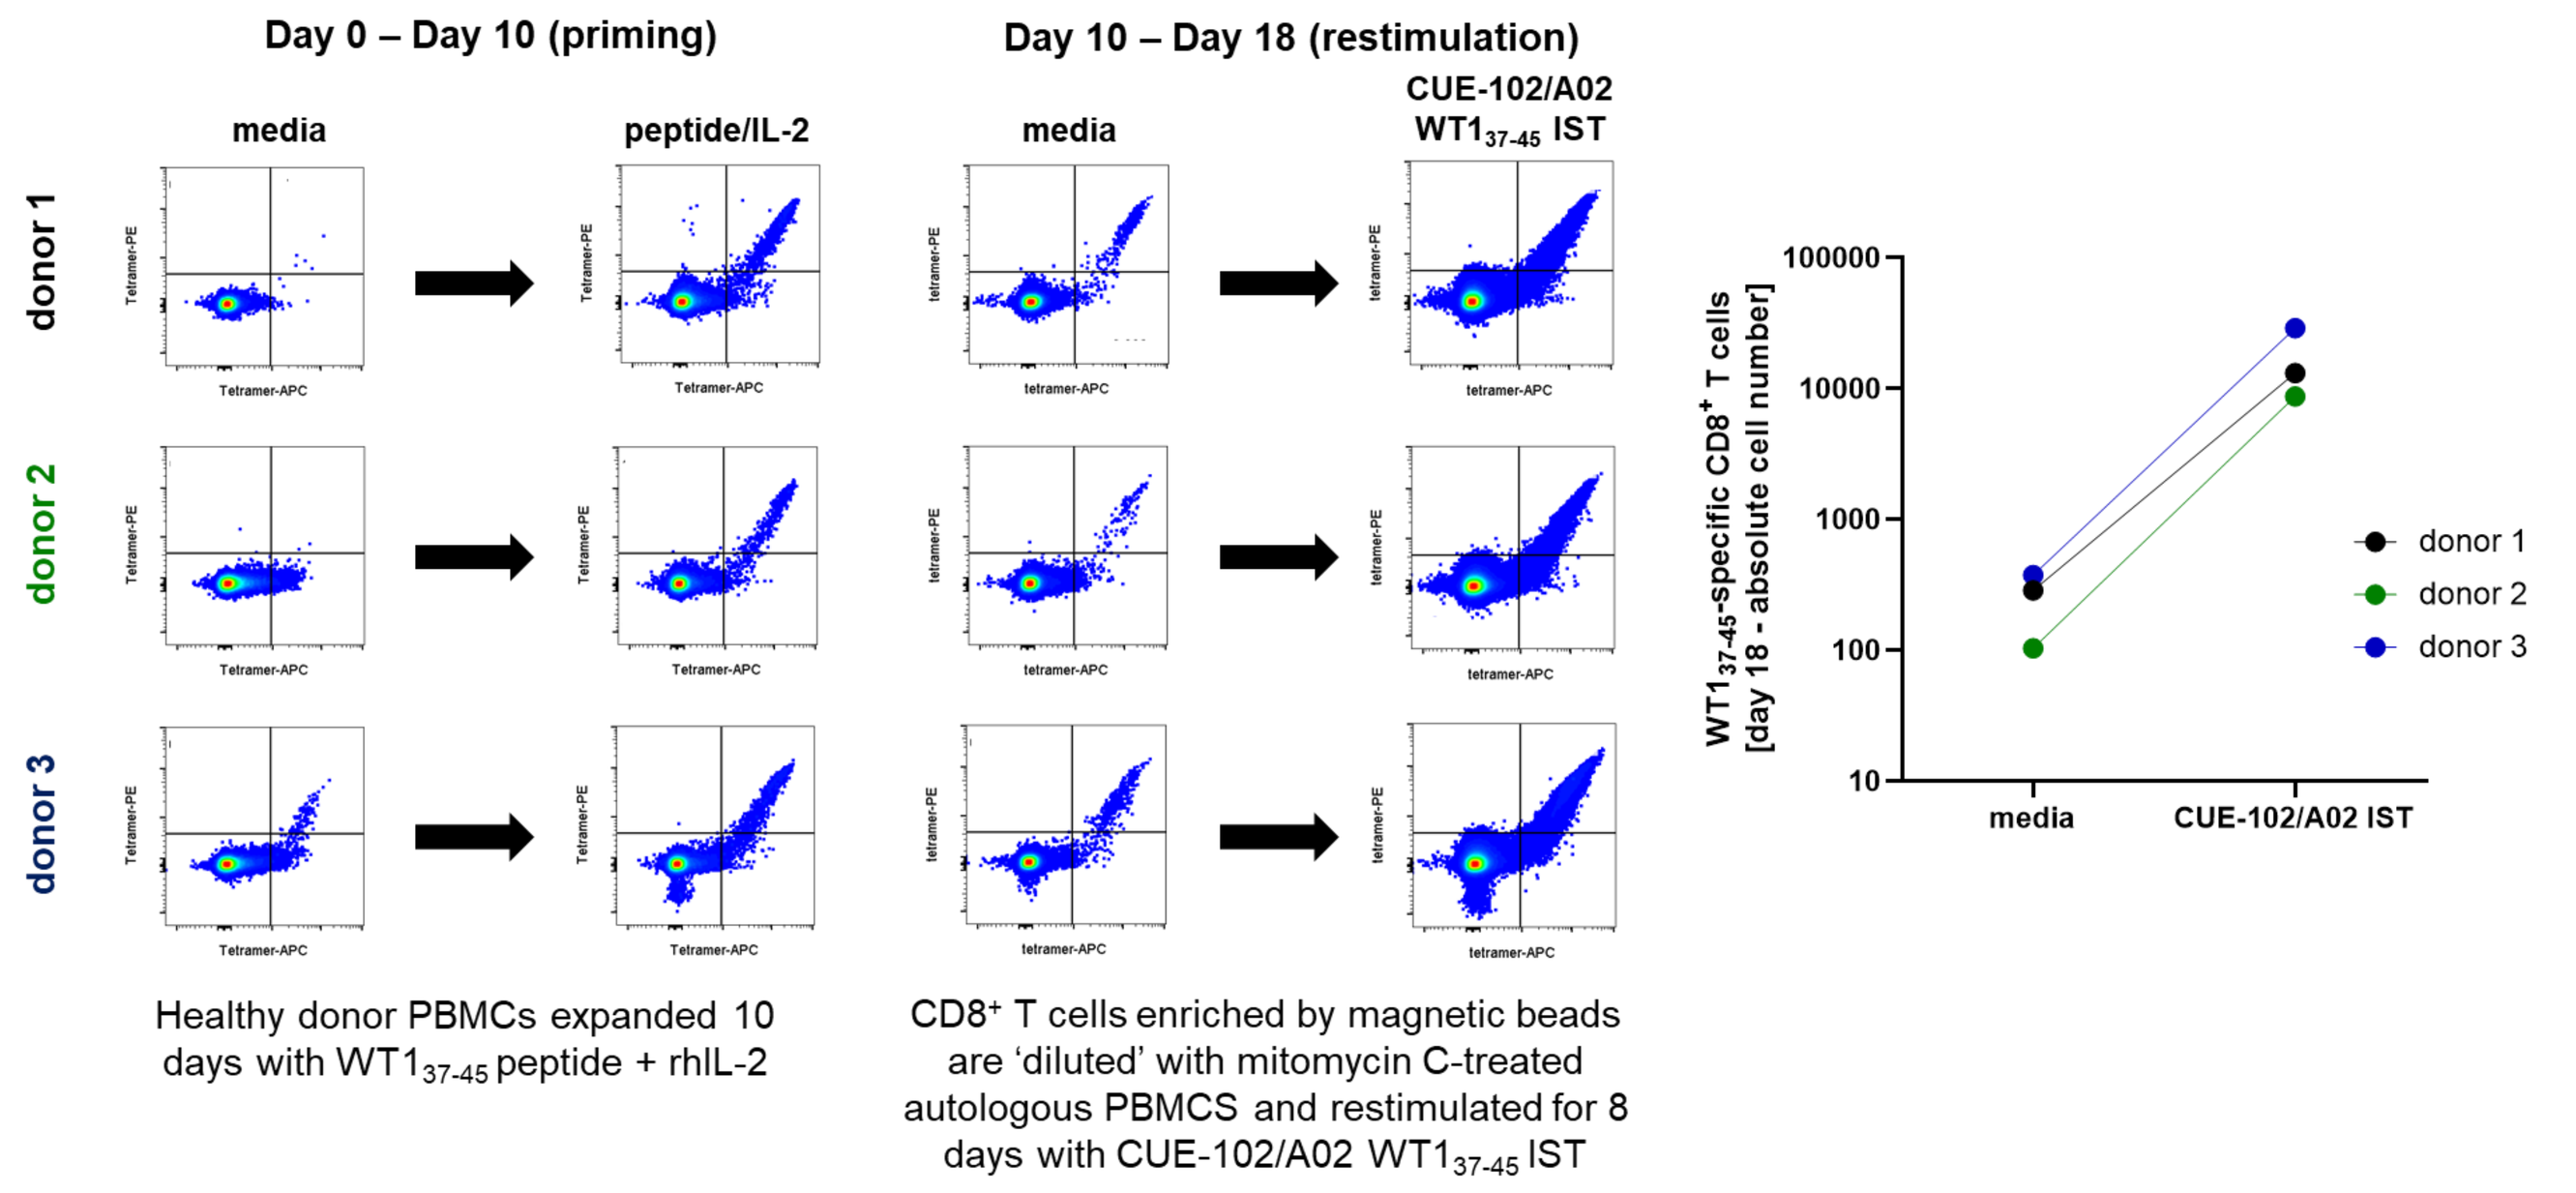

Day 0 – Day 10 (priming)
Day 10 – Day 18 (restimulation)
media
peptide/IL-2
media
CUE-102/A02 WT1 37-45 IST
donor 1
donor 2
donor 3
Tetramer-PE
Tetramer-APC
WT1 37-45-specific CD8+ T cells [day 18 - absolute cell number]
100000
10000
1000
100
10
media
CUE-102/A02 IST
donor 1
donor 2
donor 3

Healthy donor PBMCs expanded 10 days with $WT1_{37-45}$ peptide + rhIL-2

$CD8^+$ T cells enriched by magnetic beads are 'diluted' with mitomycin C-treated autologous PBMCS and restimulated for 8 days with CUE-102/A02 $WT1_{37-45}$ IST

Healthy donor PBMCs were primed for 10 days with $WT1_{37-45}$ peptide in the presence of recombinant human IL-2. $CD8^+$ T cells were then enriched by magnetic separation and restimulated with the CUE-102/A02 Immuno-STAT in ImmunocultTM media in the presence of mitomycin C-treated autologous PBMCs for 8 days. Unstimulated cells were used as negative control. Peptide-specific $CD8^+$ T cells were detected by flow cytometry upon staining with $WT1_{37-45}$-specific tetramers.

CUE-102/A02 vs Wild-Type IL-2: CUE's Attenuated IL-2 Mitigates the Risk Associated with Systemic IL-2 Activation

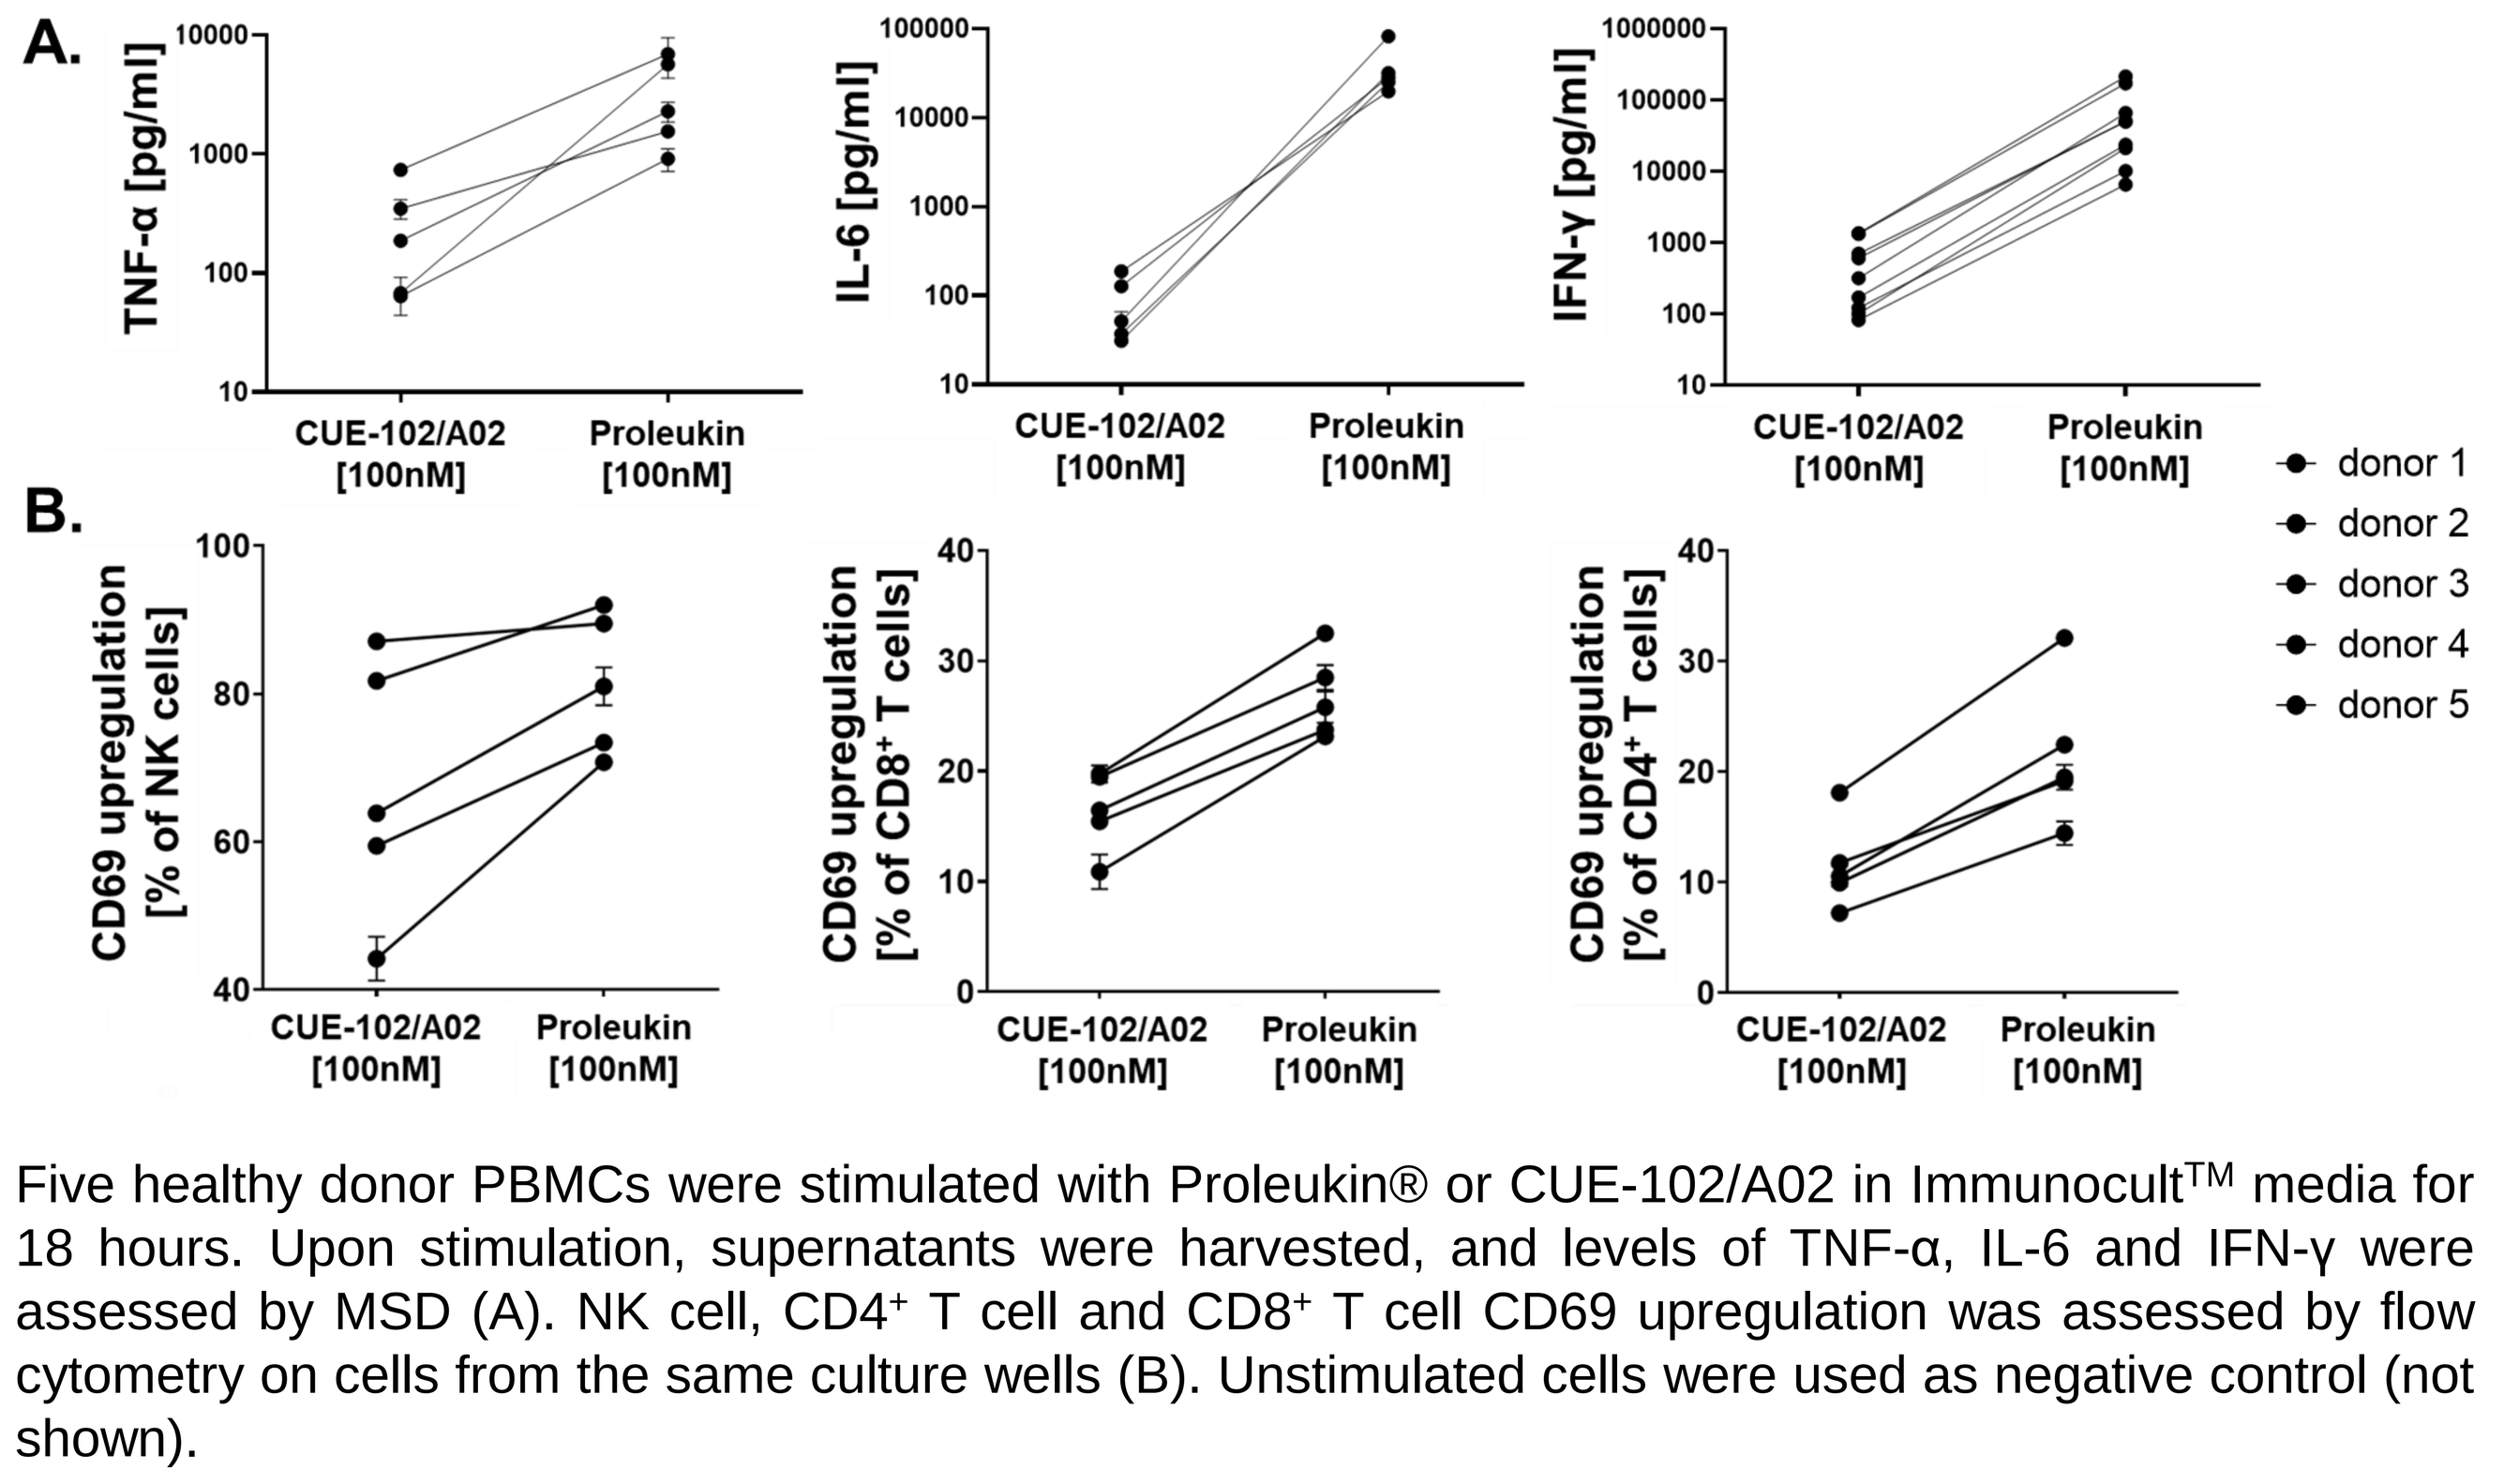

A.
TNF-α [pg/ml]
IL-6 [pg/ml]
IFN-γ [pg/ml]
CUE-102/A02 [100nM]
Proleukin [100nM]
B.
CD69 upregulation [% of NK cells]
CD69 upregulation [% of CD8+ T cells]
CD69 upregulation [% of CD4+ T cells]
donor 1
donor 2
donor 3
donor 4
donor 5

Five healthy donor PBMCs were stimulated with Proleukin® or CUE-102/A02 in ImmunocultTM media for 18 hours. Upon stimulation, supernatants were harvested, and levels of TNF-α, IL-6 and IFN-γ were assessed by MSD (A). NK cell, $CD4^+$ T cell and $CD8^+$ T cell CD69 upregulation was assessed by flow cytometry on cells from the same culture wells (B). Unstimulated cells were used as negative control (not shown).

CUE-102/A02 Expanded T Cells are Polyfunctional CTLs that Recognize and Kill $WT1_{37-45}$ Peptide-Presenting Target Cells

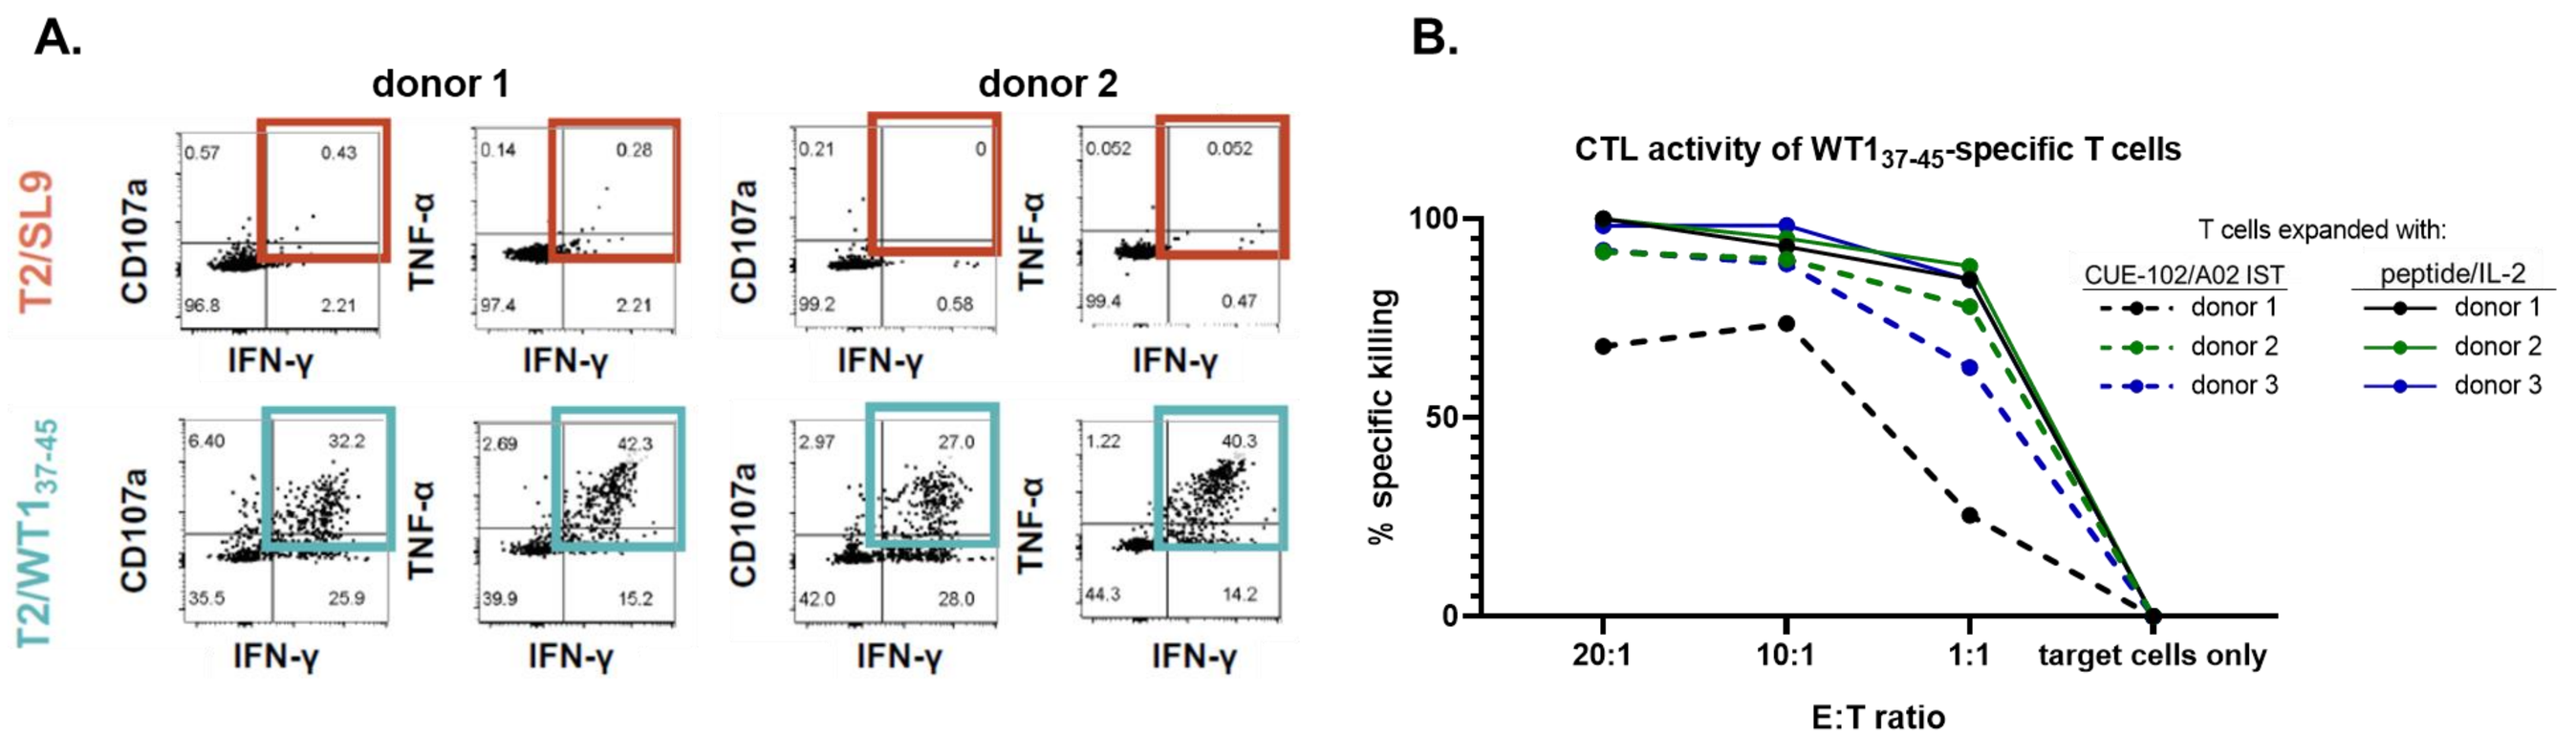

A.
donor 1
donor 2
T2/SL9
T2/WT1 37-45
CD107a
TNF-α
IFN-γ
B.
CTL activity of WT1 37-45-specific T cells
T cells expanded with:
CUE-102/A02 IST
peptide/IL-2
donor 1
donor 2
donor 3
% specific killing
20:1
10:1
1:1
target cells only
E:T ratio

Healthy donor PBMCs were primed for 10 days with $WT1_{37-45}$ peptide in the presence of recombinant human IL-2 and expanded for 8 days with $WT1_{37-45}$ peptide or with the CUE-102/A02 $WT1_{37-45}$ Immuno-STAT in ImmunocultTM media in the presence of mitomycin C-treated autologous PBMCs. $WT1_{37-45}$-specific $CD8^+$ T cells were enriched by magnetic bead-based separation using $WT1_{37-45}$-specific PE-labeled tetramers. (A) CUE102/A02 IST-expanded $WT1_{37-45}$-specific T cells express effector molecules IFN-γ, TNF-α, and upregulate the degranulation marker CD107a upon 4 hours of interaction with target T2 cells pulsed with the cognate $WT1_{37-45}$ peptide but not with an irrelevant peptide. (B) Expanded $WT1_{37-45}$-specific T cells kill cognate $WT1_{37-45}$ peptide-pulsed T2 cells but not control peptide-pulsed T2 cells in overnight cultures performed at different T cell effector:target cell ratios. Specific killing is assessed by flow cytometry comparing the ratio of viable T2 cell pulsed with cognate vs control upon overnight culture.

Harnessing platform modularity to develop Immuno-STATs targeting patients of different HLA haplotypes

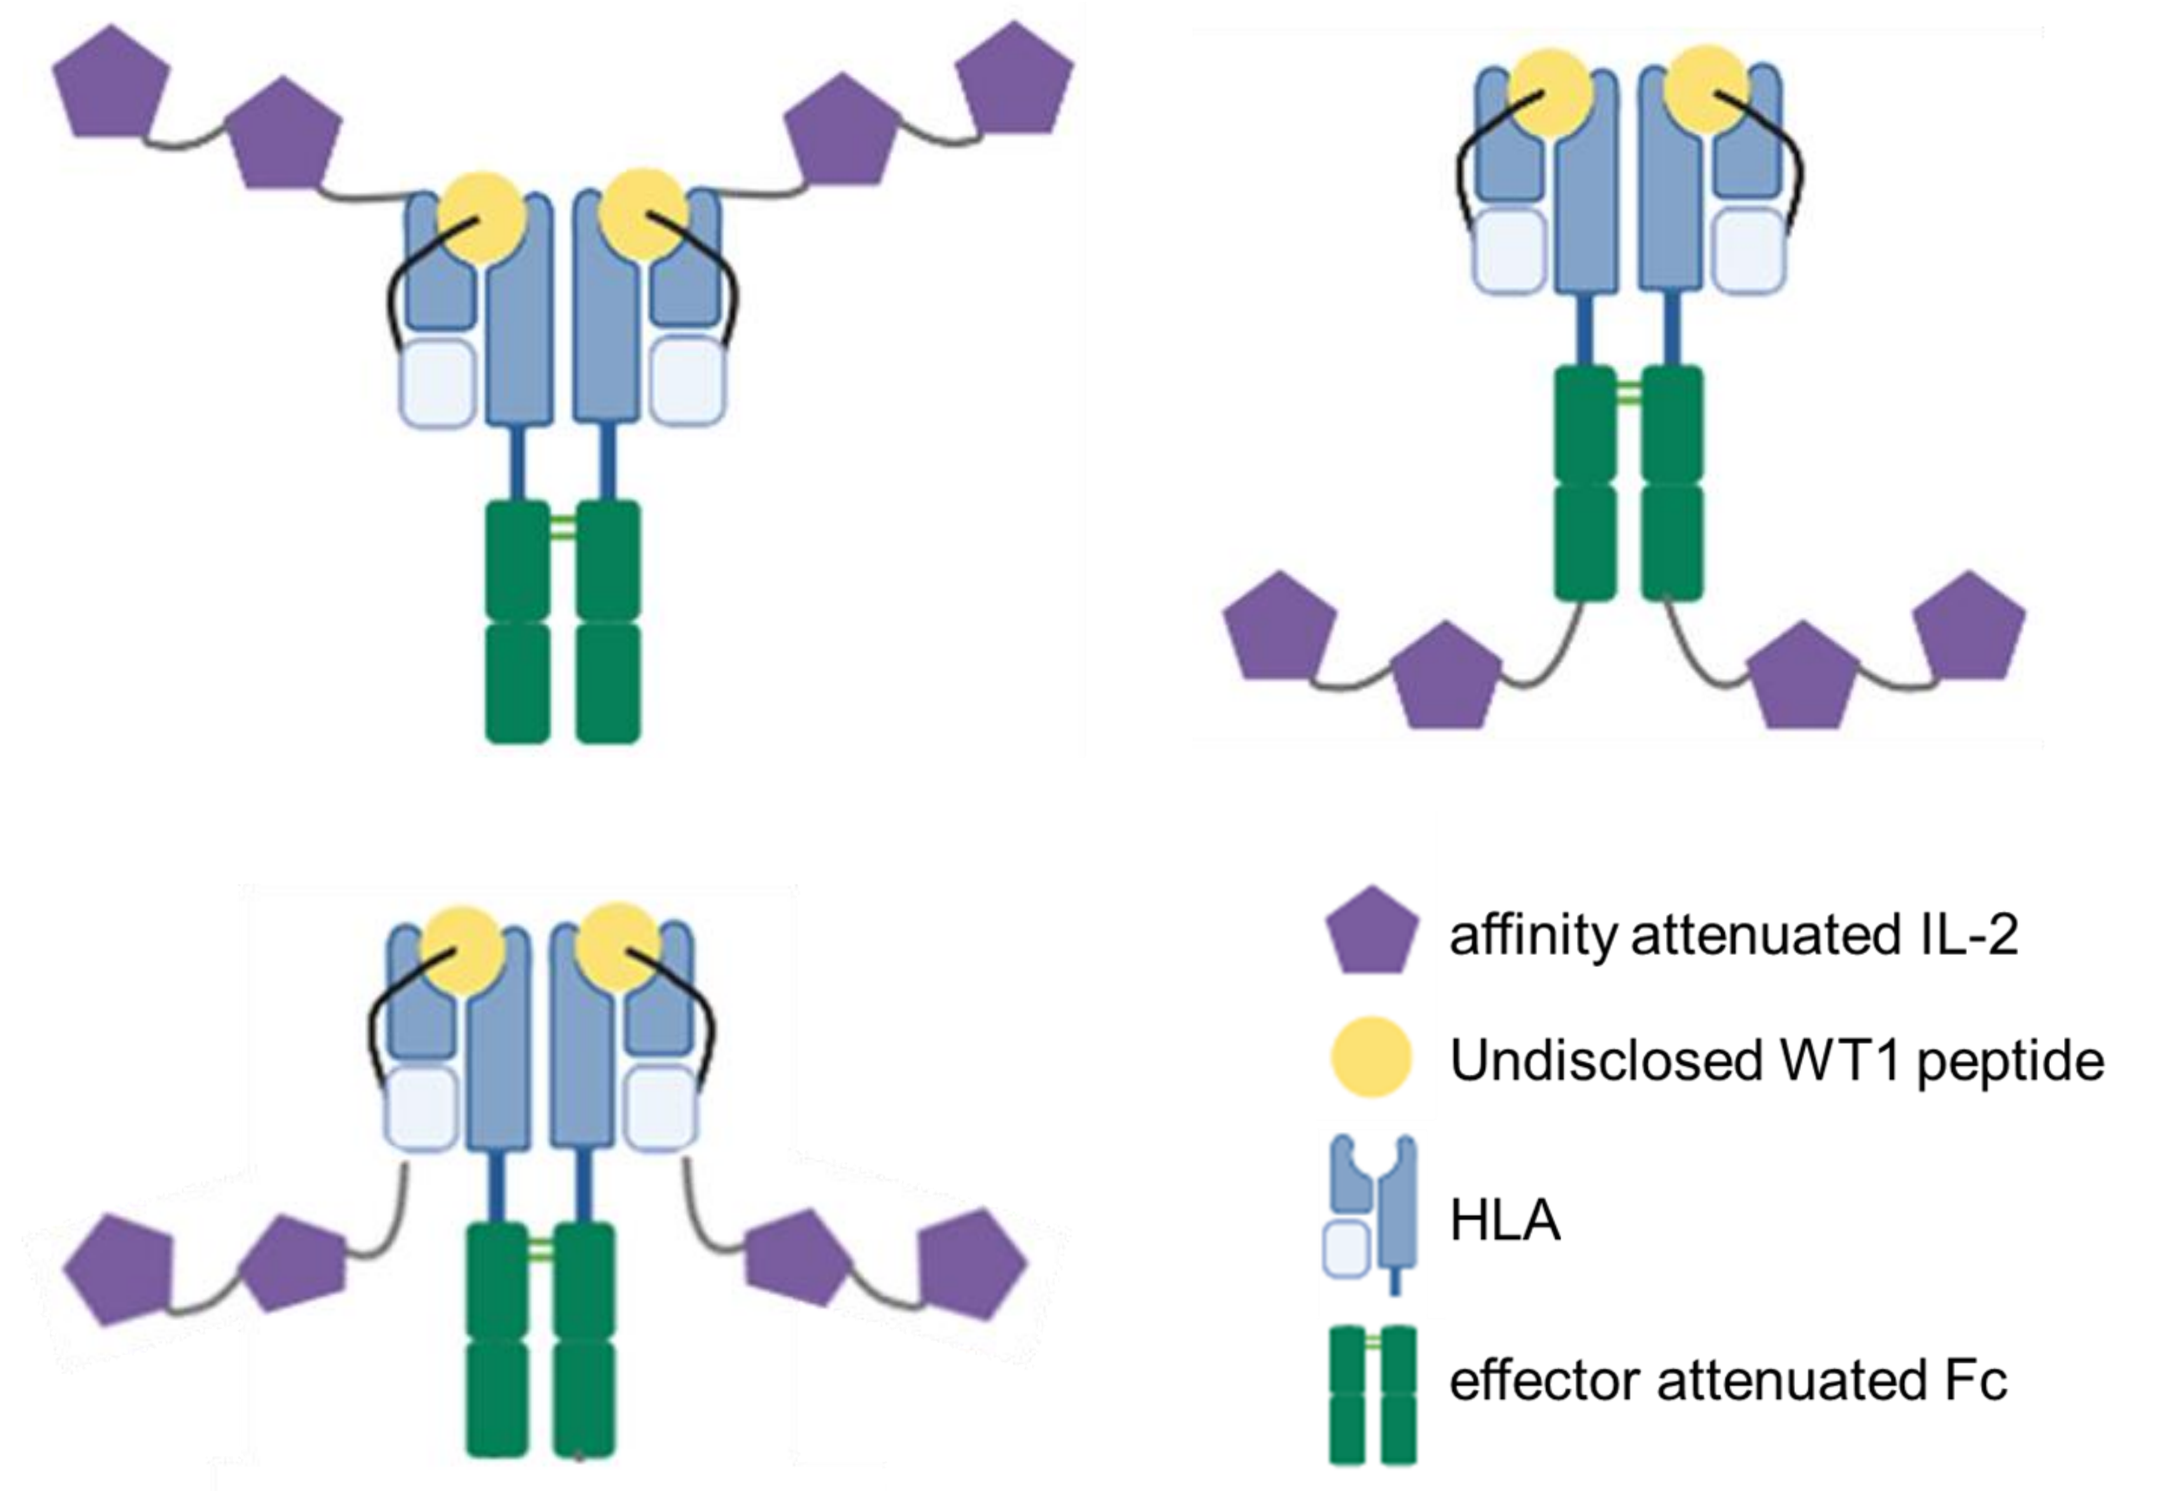

affinity attenuated IL-2
Undisclosed WT1 peptide
HLA
effector attenuated Fc

Immuno-STATs targeting patients of different HLA haplotypes were designed and various Immuno-STAT frameworks, such as those shown above having different epitope candidates and different HLA molecules, were analyzed. Our analysis considered numerous factors specific to the combinations of the framework, HLA allele and potential epitopes, including manufacturability, protein stability and bioactivity.

Immuno-STAT modularity enables platform expansion to HLA-A11⁺ and HLA-A24⁺ patients across tumor indications associated with WT1 and KRAS

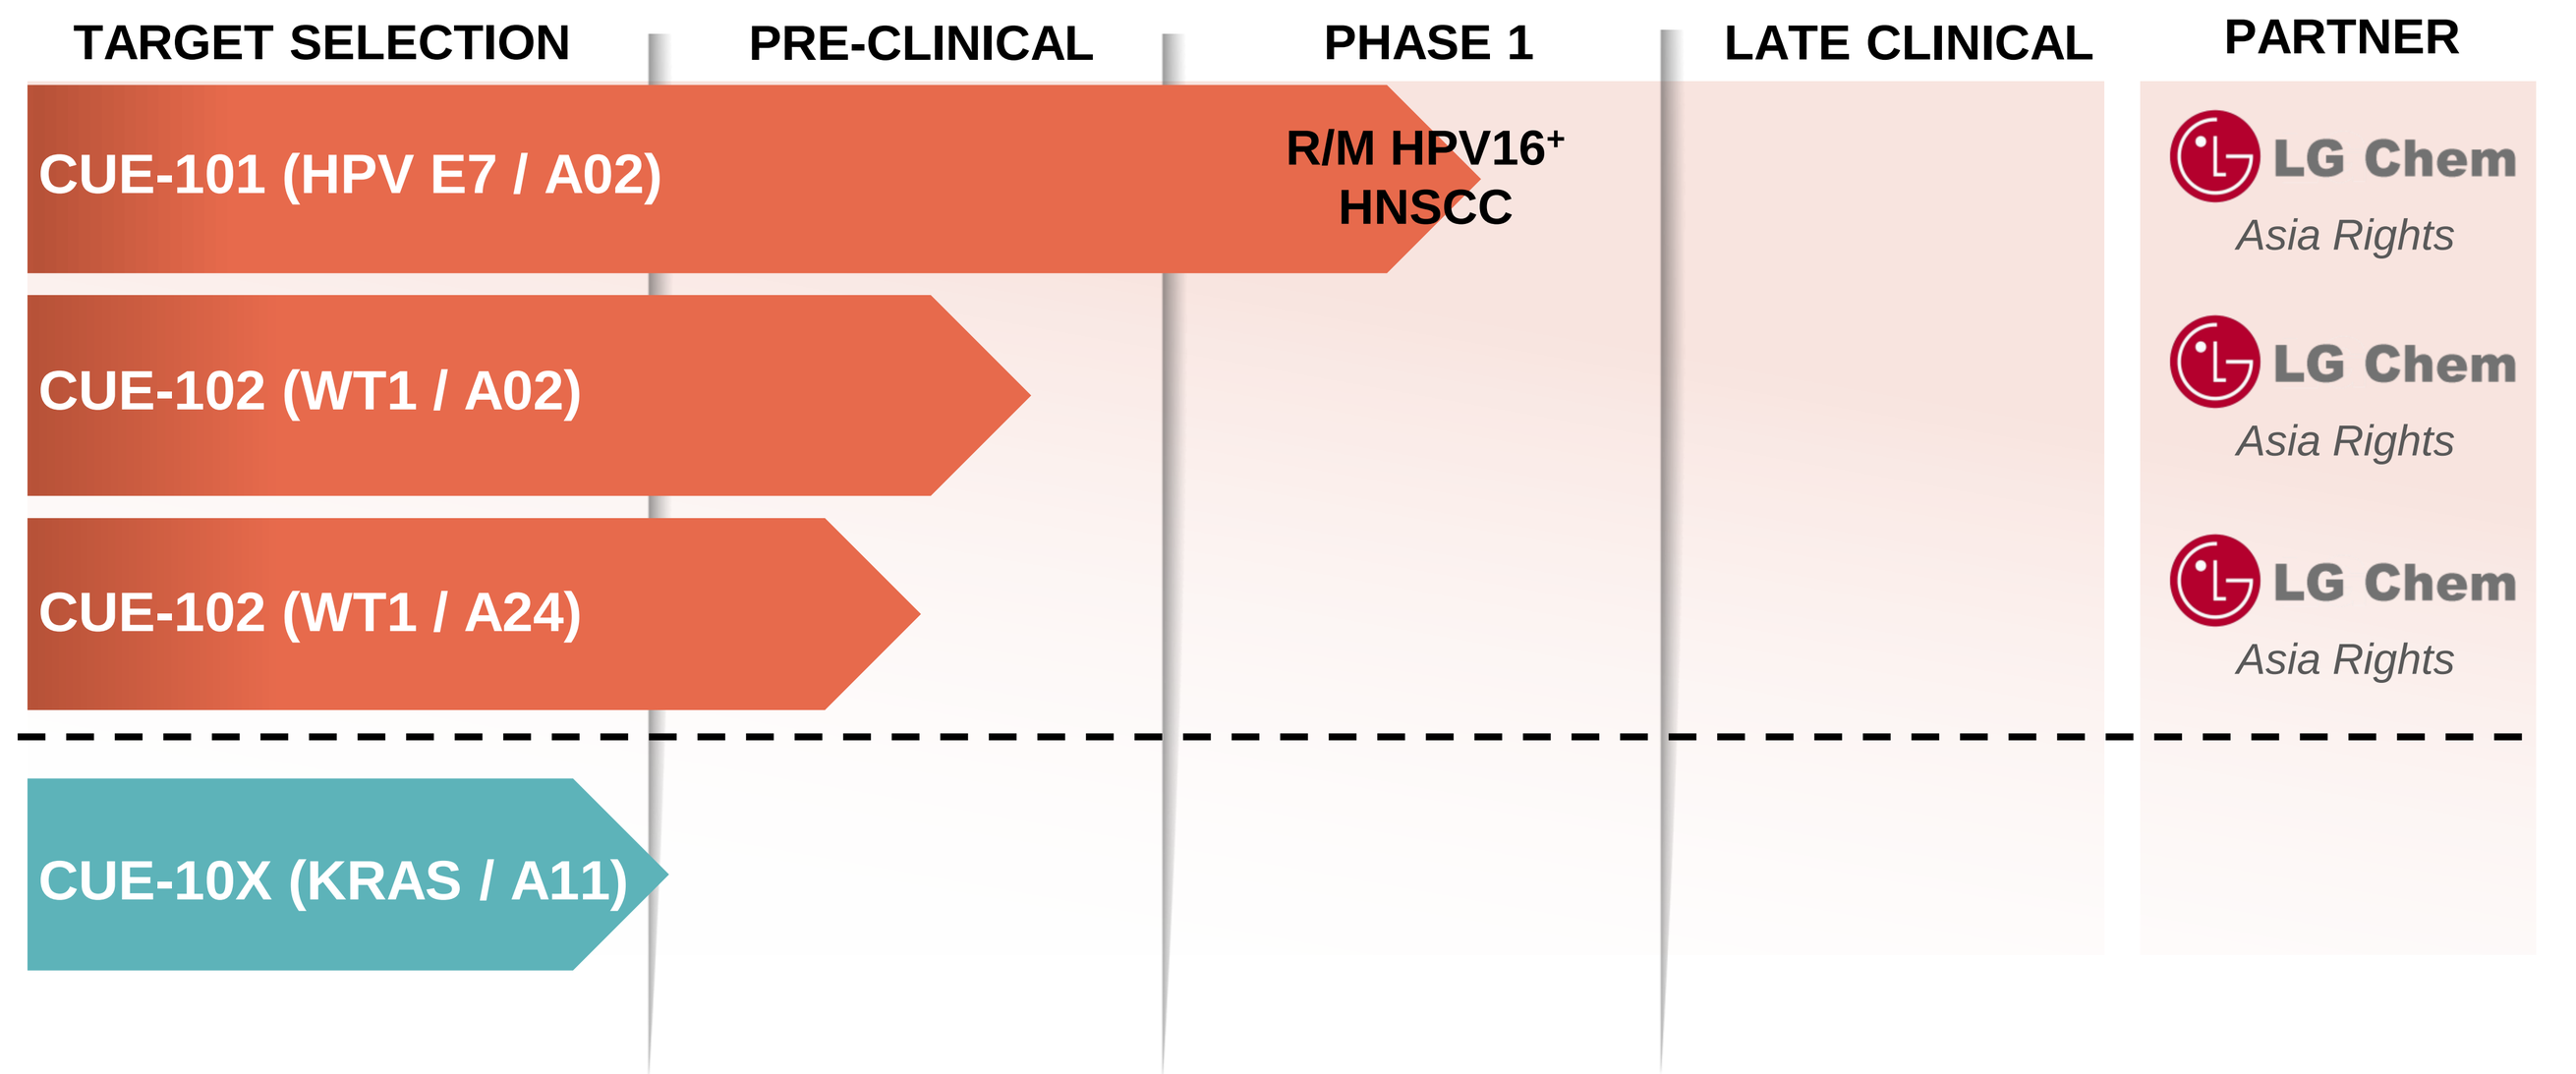

TARGET SELECTION
PRE-CLINICAL
PHASE 1
LATE CLINICAL
PARTNER
CUE-101 (HPV E7 / A02)
R/M HPV16+ HNSCC
LG Chem
Asia Rights
CUE-102 (WT1 / A02)
LG Chem
Asia Rights
CUE-102 (WT1 / A24)
LG Chem
Asia Rights
CUE-10X (KRAS / A11)

Conclusions

In partnership with LG Chem, we have developed novel Immuno-STAT fusion proteins that are comprised of HLA-A*02 or HLA-A*24 molecules presenting peptide epitopes derived from WT1, four copies of affinity-attenuated human IL-2, and an effector attenuated human IgG1 Fc domain. CUE-102 molecules induce expansion of WT1-specific $CD8^+$ T cells from both unprimed and peptide-primed PBMC. The repertoire of the CUE-102-expanded $CD8^+$ T cells, their polyfunctionality and ability to recognize and respond to WT1 peptide-presenting target cells suggest that CUE-102 Immuno-STATs have the potential to enhance anti-tumor immunity in patients with WT1-positive malignancies.
As exemplary of the platform and the IL-2-based CUE-100 series, our first molecule CUE-101 is currently being evaluated in a phase I trial in recurrent-metastatic HPV-driven HNSCC.

QR code

Presented at AACR Virtual Annual Meeting II • June 22-24, 2020

© 2020 Cue Biopharma. All rights reserved.